P2.09-13

QT-interval prolongation and Torsades de Pointes with EGFR TKIs in NSCLC: targeted literature review

Bilal Khokhar,[1] Beatrice Chiang,[1] Kristy Iglay,[2] Kamika Reynolds,[2,3] Nidia Rodriguez-Ormaza,[2,3] William Spalding,[1] Eric Freedland[1]

[1]Takeda Development Center Americas, Inc., Lexington, MA, USA; [2]CERobs Consulting, LLC, Wrightsville Beach, NC, USA; [3]University of North Carolina at Chapel Hill, Chapel Hill, NC, USA

Background

- Changes in heart rhythm, such as QT-interval prolongation (QTp) or Torsades de Pointes (TdP), are serious conditions that can lead to sudden death if not recognized and treated[1]
- QTp and TdP can occur with some cancer treatments, even in patients without relevant clinical history[2]
 - In particular, QTp has been reported with some epidermal growth factor receptor (EGFR) tyrosine kinase inhibitors (TKIs) in patients with non–small cell lung cancer (NSCLC)[3,4]
- This targeted literature review aimed to provide a foundational understanding of the risks of QTp and TdP associated with EGFR TKIs in patients with NSCLC

Methods

- A targeted literature review was conducted to determine the incidence and mortality of QTp and TdP in patients with NSCLC who received EGFR TKIs
 - An electronic search and a manual search were conducted
- Incidence of and mortality due to QTp and TdP were outcomes of interest
- Populations assessed were patients with any cancer type treated with TKIs and patients with NSCLC treated with EGFR TKIs
- Specific EGFR TKIs of interest included in the search were afatinib, almonertinib, dacomitinib, erlotinib, gefitinib, icotinib, mobocertinib, olmutinib, osimertinib, and simotinib
- Publication types included single-arm studies, studies with multiple arms of a single drug under examination, comparative studies versus patients with lung cancer treated with prespecified non-TKIs, and case reports

Results

- A total of 296 publications were identified, of which 86 met all eligibility criteria and were abstracted for incidence or mortality due to QTp and TdP **(Figure 1)**
 - For patients with any type of cancer treated with TKIs, 4 studies reported data on the incidence of or mortality due to QTp and/or TdP
 - For patients with NSCLC treated with EGFR TKIs, 83 publications reported incidence of or mortality due to QTp and/or TdP
 - No studies on almonertinib met eligibility criteria

Figure 1: PRISMA flow diagram for systematic review

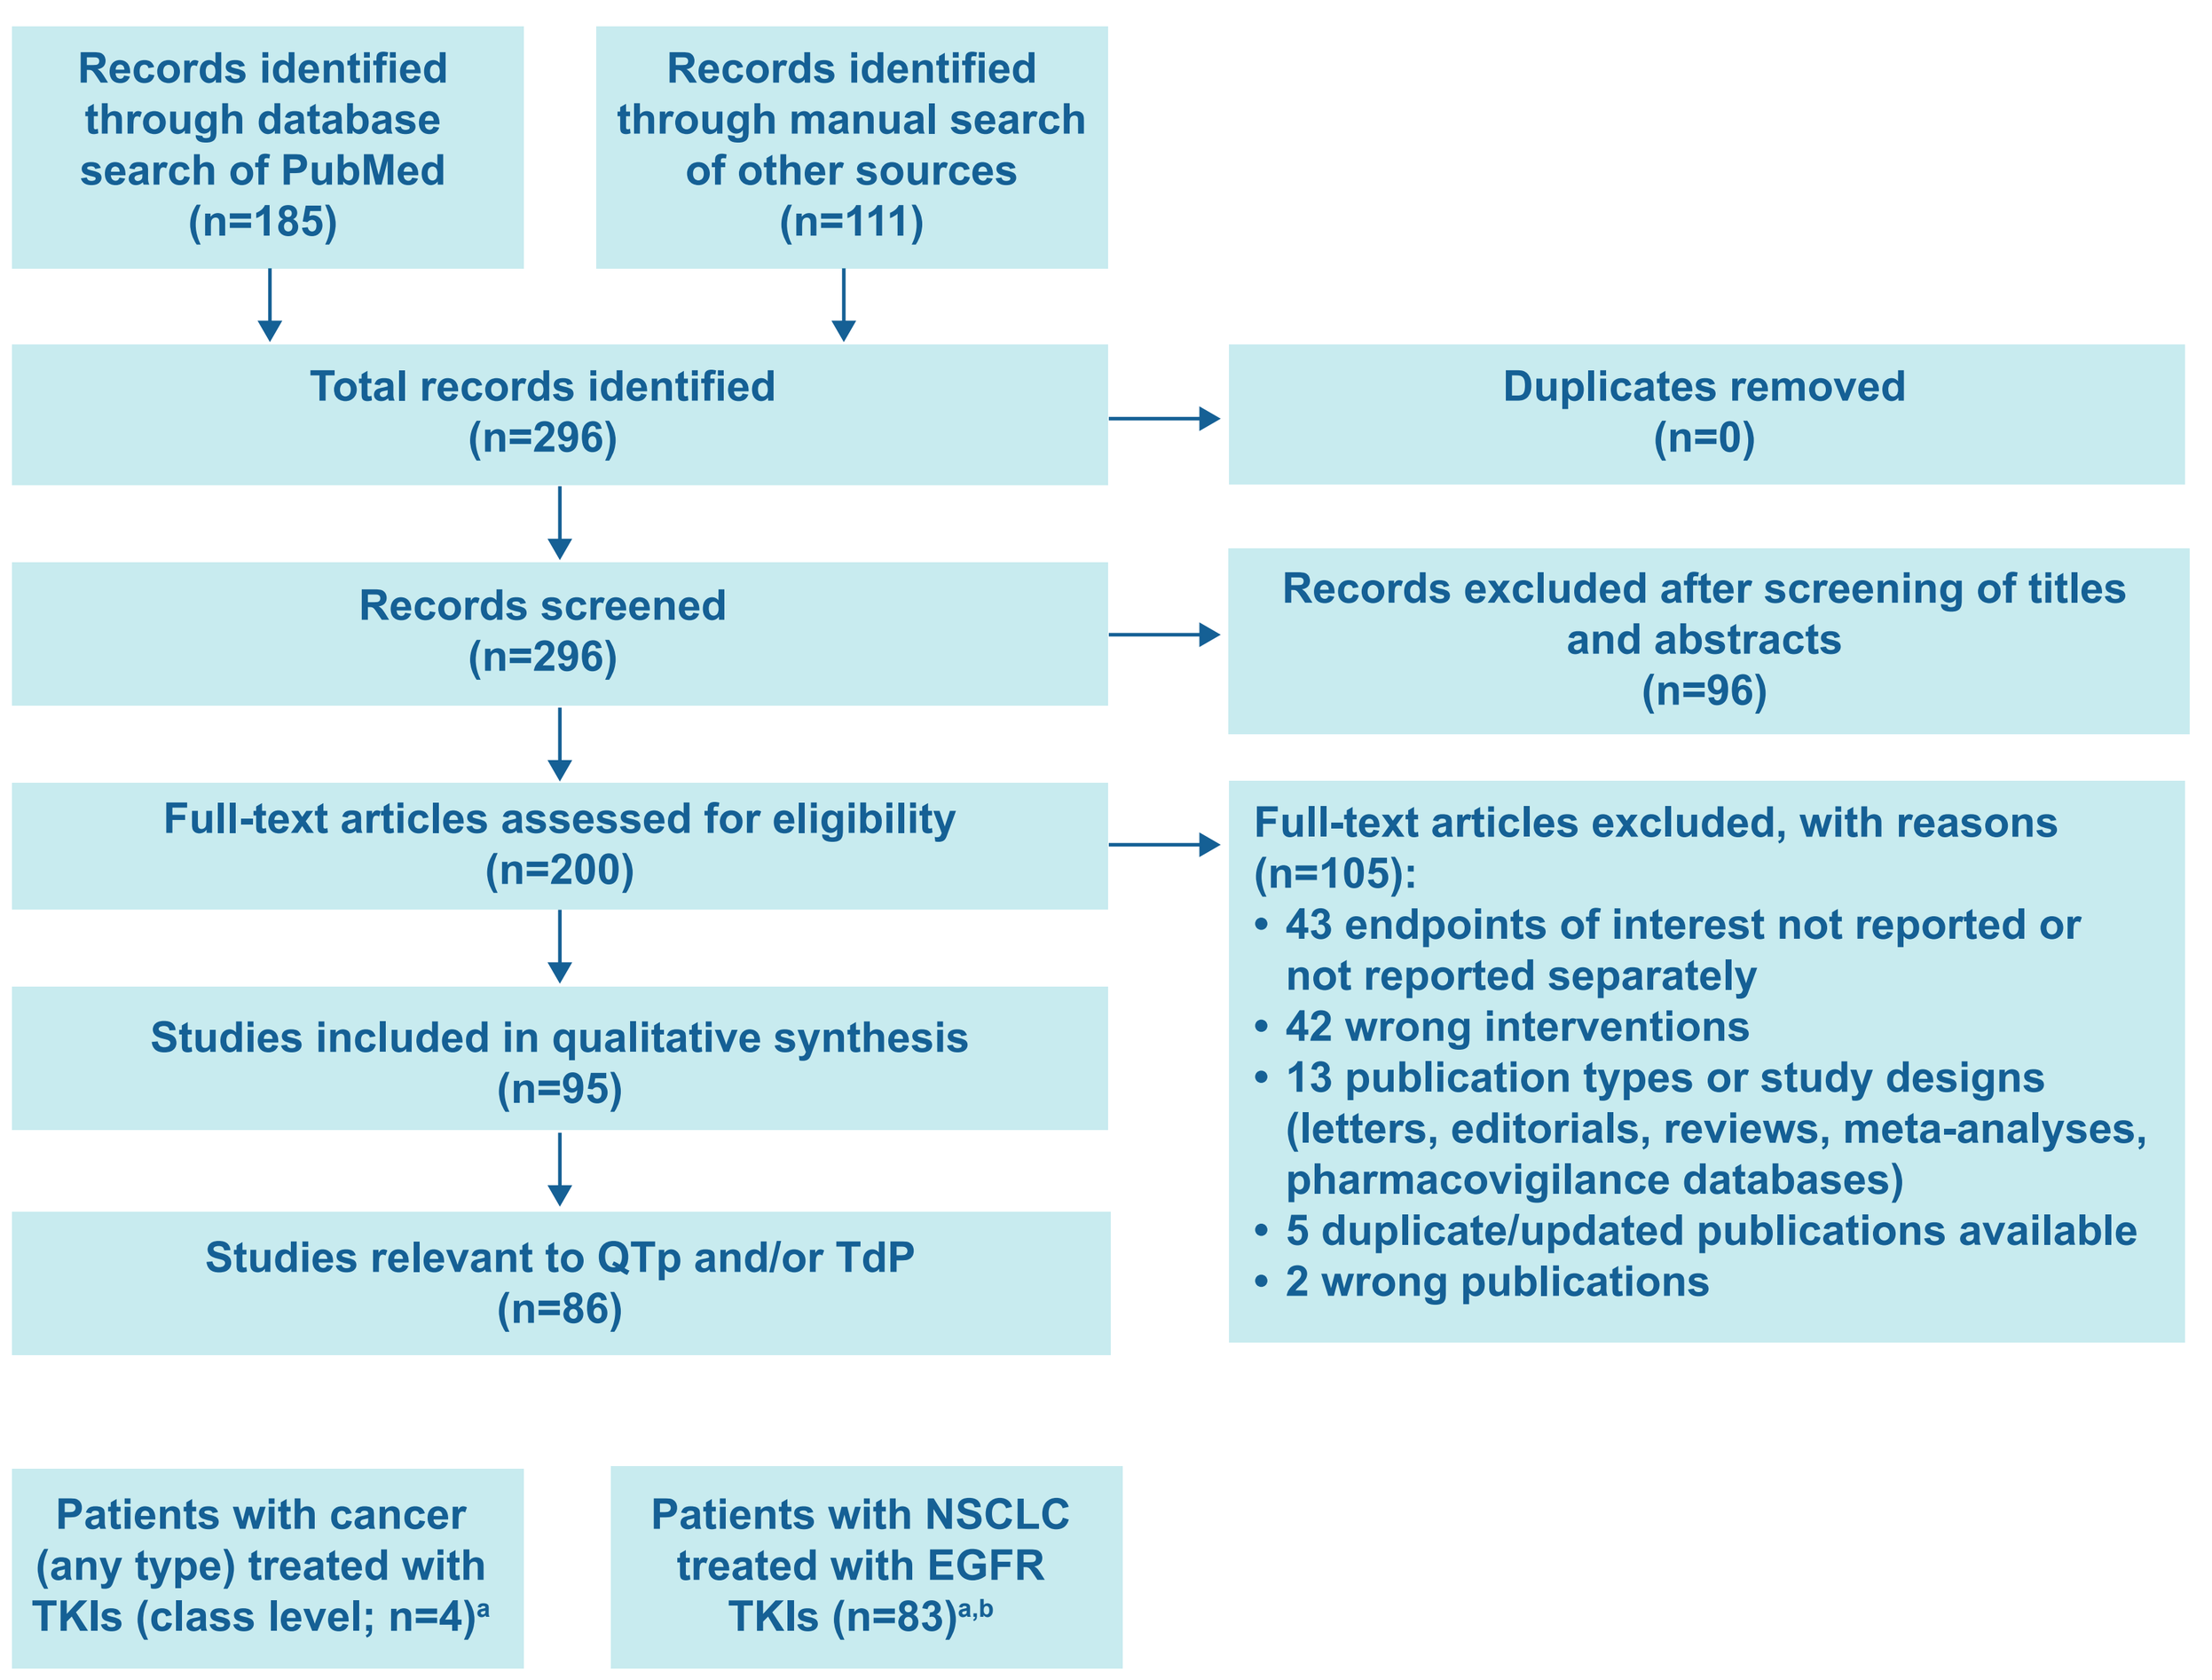
Records identified through database search of PubMed (n=185)
Records identified through manual search of other sources (n=111)
Total records identified (n=296)
Duplicates removed (n=0)
Records screened (n=296)
Records excluded after screening of titles and abstracts (n=96)
Full-text articles assessed for eligibility (n=200)
Full-text articles excluded, with reasons (n=105):
• 43 endpoints of interest not reported or not reported separately
• 42 wrong interventions
• 13 publication types or study designs (letters, editorials, reviews, meta-analyses, pharmacovigilance databases)
• 5 duplicate/updated publications available
• 2 wrong publications
Studies included in qualitative synthesis (n=95)
Studies relevant to QTp and/or TdP (n=86)
Patients with cancer (any type) treated with TKIs (class level; n=4)[a]
Patients with NSCLC treated with EGFR TKIs (n=83)[a,b]

[a]1 article is included in both counts; [b]5 of these publications are case studies

- For patients with any type of cancer treated with TKIs **(Figures 2 and 3)**

QTp incidence	• In 4 studies, incidence ranged from 0% to 33.7%
QTp mortality	• In 1 study, mortality rate due to QTp was 0.3%
TdP incidence	• In 1 study, TdP incidence was 0.2%
TdP mortality	• No deaths due to TdP were reported in any studies

- For patients with NSCLC treated with EGFR TKIs **(Figures 3 and 4)**

QTp incidence	• Among 49 publications, incidence ranged from 0% to 27.8% in observational studies and 0% to 11% in noncomparative clinical trials – In comparative clinical trials, incidence ranged from 0% to 10% in EGFR TKI arms and 0% to 1% in chemotherapy arms
QTp mortality	• Mortality due to QTp was not reported in any studies
TdP incidence	• No incidences of TdP were reported in any studies[a]
TdP mortality	• No deaths were reported in any studies; 1 case report described a TdP-related death after osimertinib therapy with concomitant moxifloxacin

[a]NCI-CTCAE versions 3, 4, and 5 define TdP as a grade 4 QTp event, and the terminology varied; thus, it is possible that TdP events were not described separately by investigators.

Figure 2: Patients with any type of cancer treated with TKIs

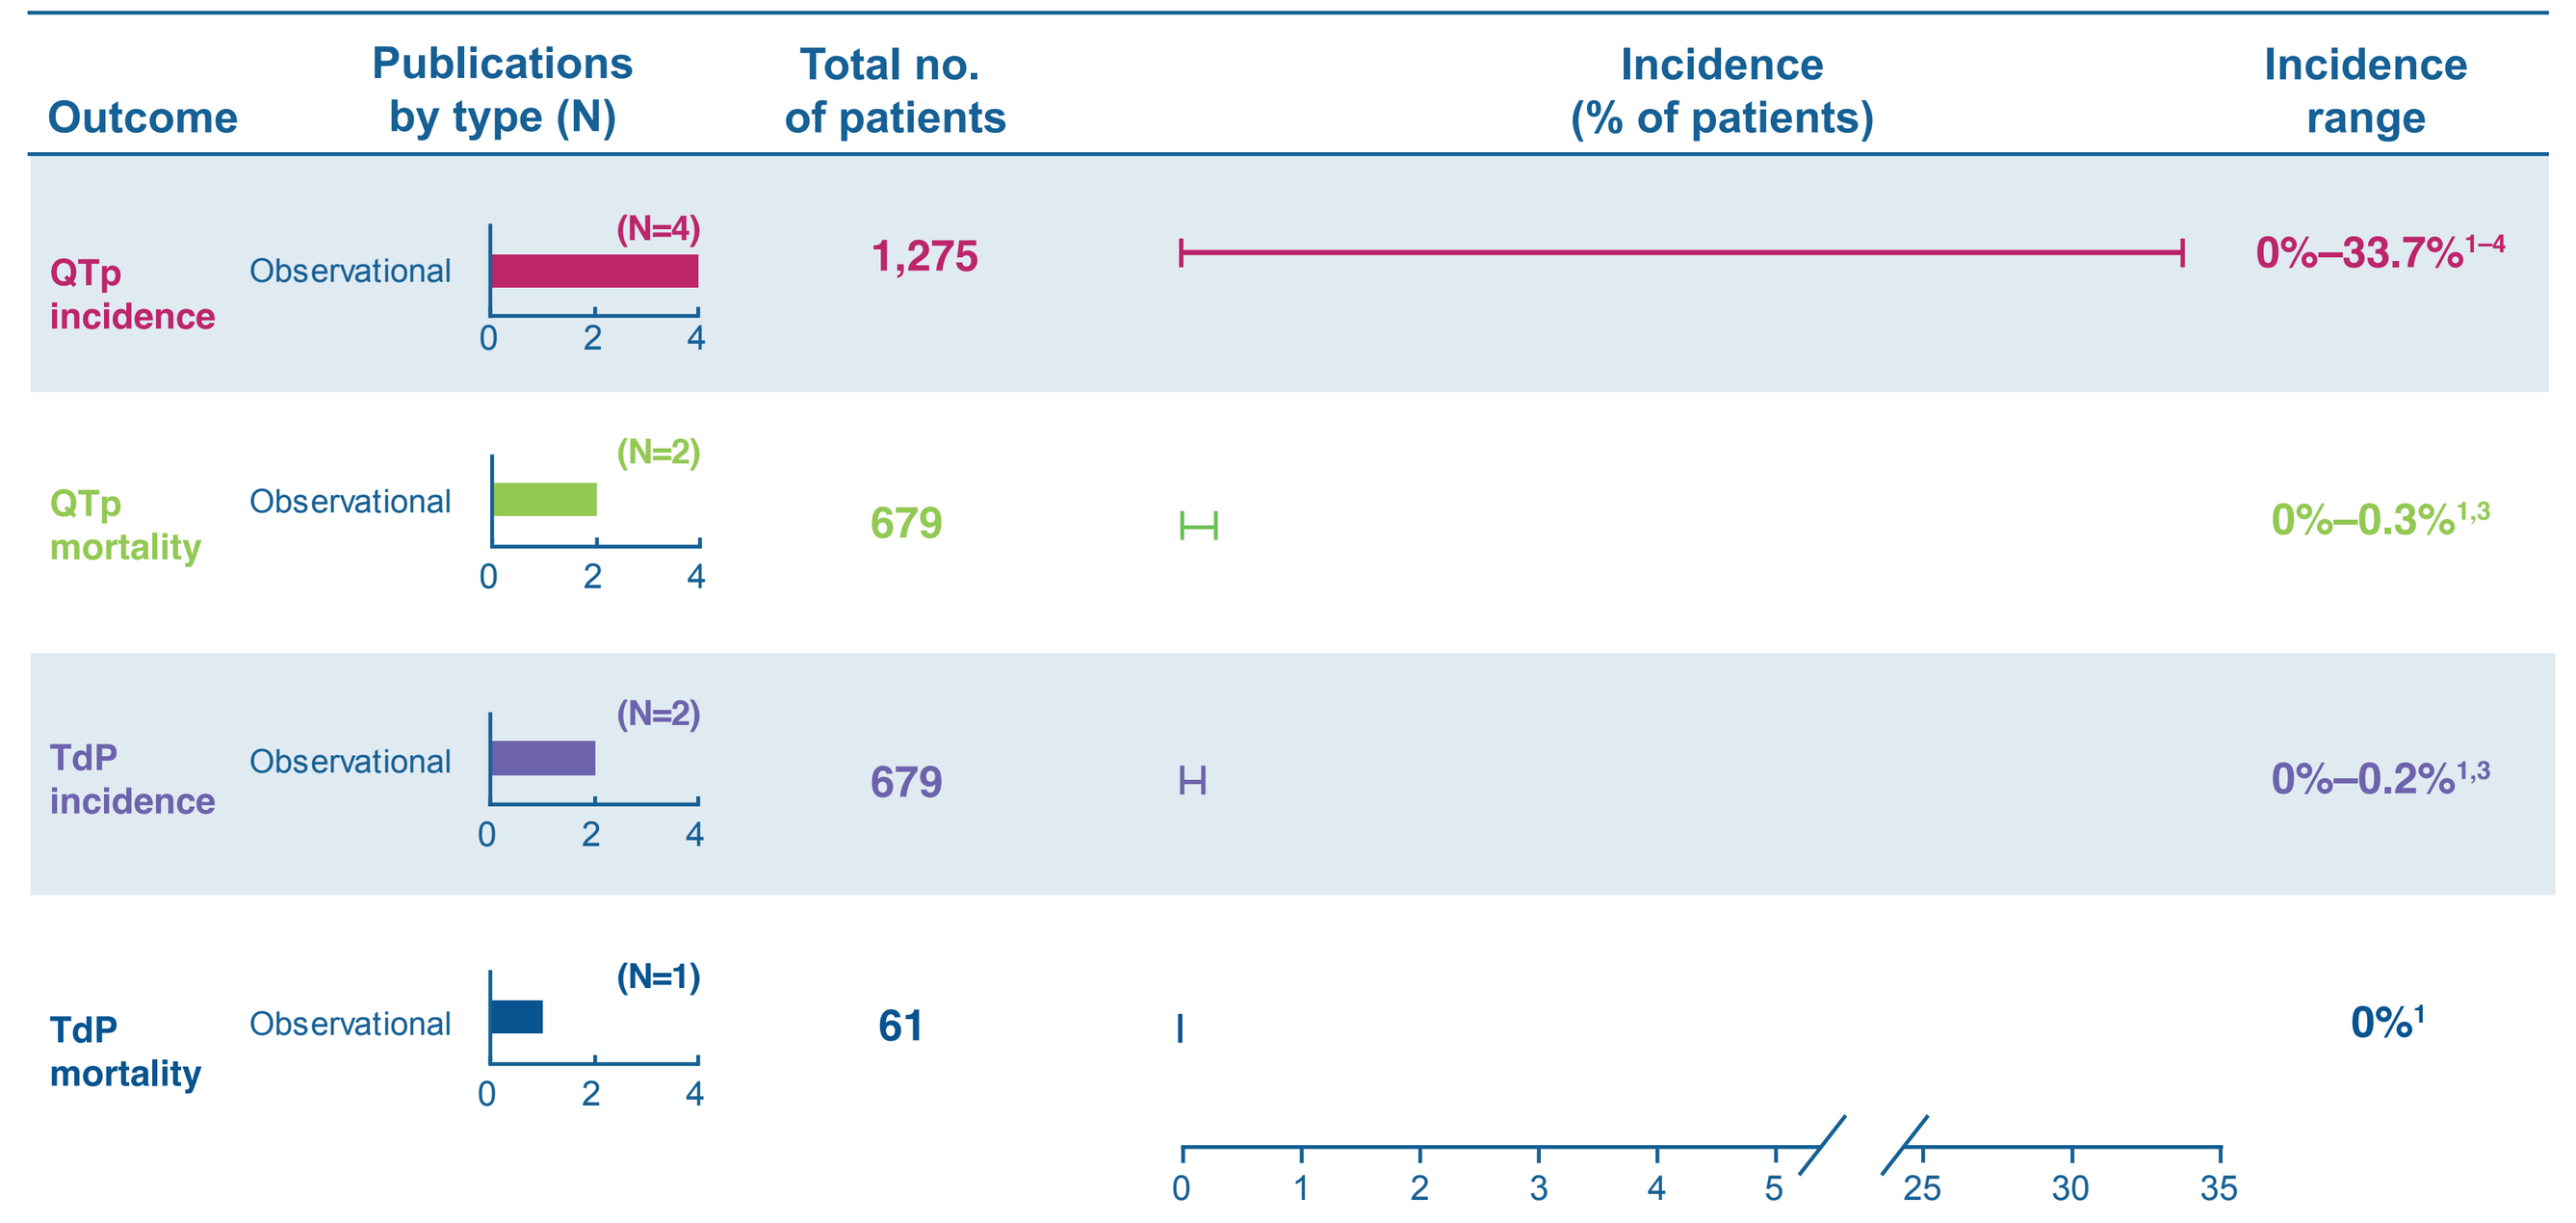
Outcome
Publications by type (N)
Total no. of patients
Incidence (% of patients)
Incidence range
QTp incidence
Observational (N=4)
1,275
0%–33.7%[1–4]
QTp mortality
Observational (N=2)
679
0%–0.3%[1,3]
TdP incidence
Observational (N=2)
679
0%–0.2%[1,3]
TdP mortality
Observational (N=1)
61
0%[1]
0 1 2 3 4 5 25 30 35

1. Abu Rmilah AA, et al. Int J Cancer. 2020;147(11):3160–67. 2. Assunção PM, et al. Clin Lymphoma Myeloma Leuk. 2019;19(3):162–66. 3. Casavecchia G, et al. Acta Cardiol. 2022;77(2):130–35. 4. Kloth JS, et al. Br J Cancer. 2015;112(6):1011–16.

Question Does TKI treatment impact a patient's risk for QTp and/or TdP?

Investigation

86 publications relevant to QTp and/or TdP identified by systematic literature review

4 publications reporting QTp and/or TdP in patients with any cancer treated with TKIs

83 publications reporting QTp and/or TdP in patients with NSCLC treated with EGFR TKIs

Results

Figure 3: Systematic literature review—overall results

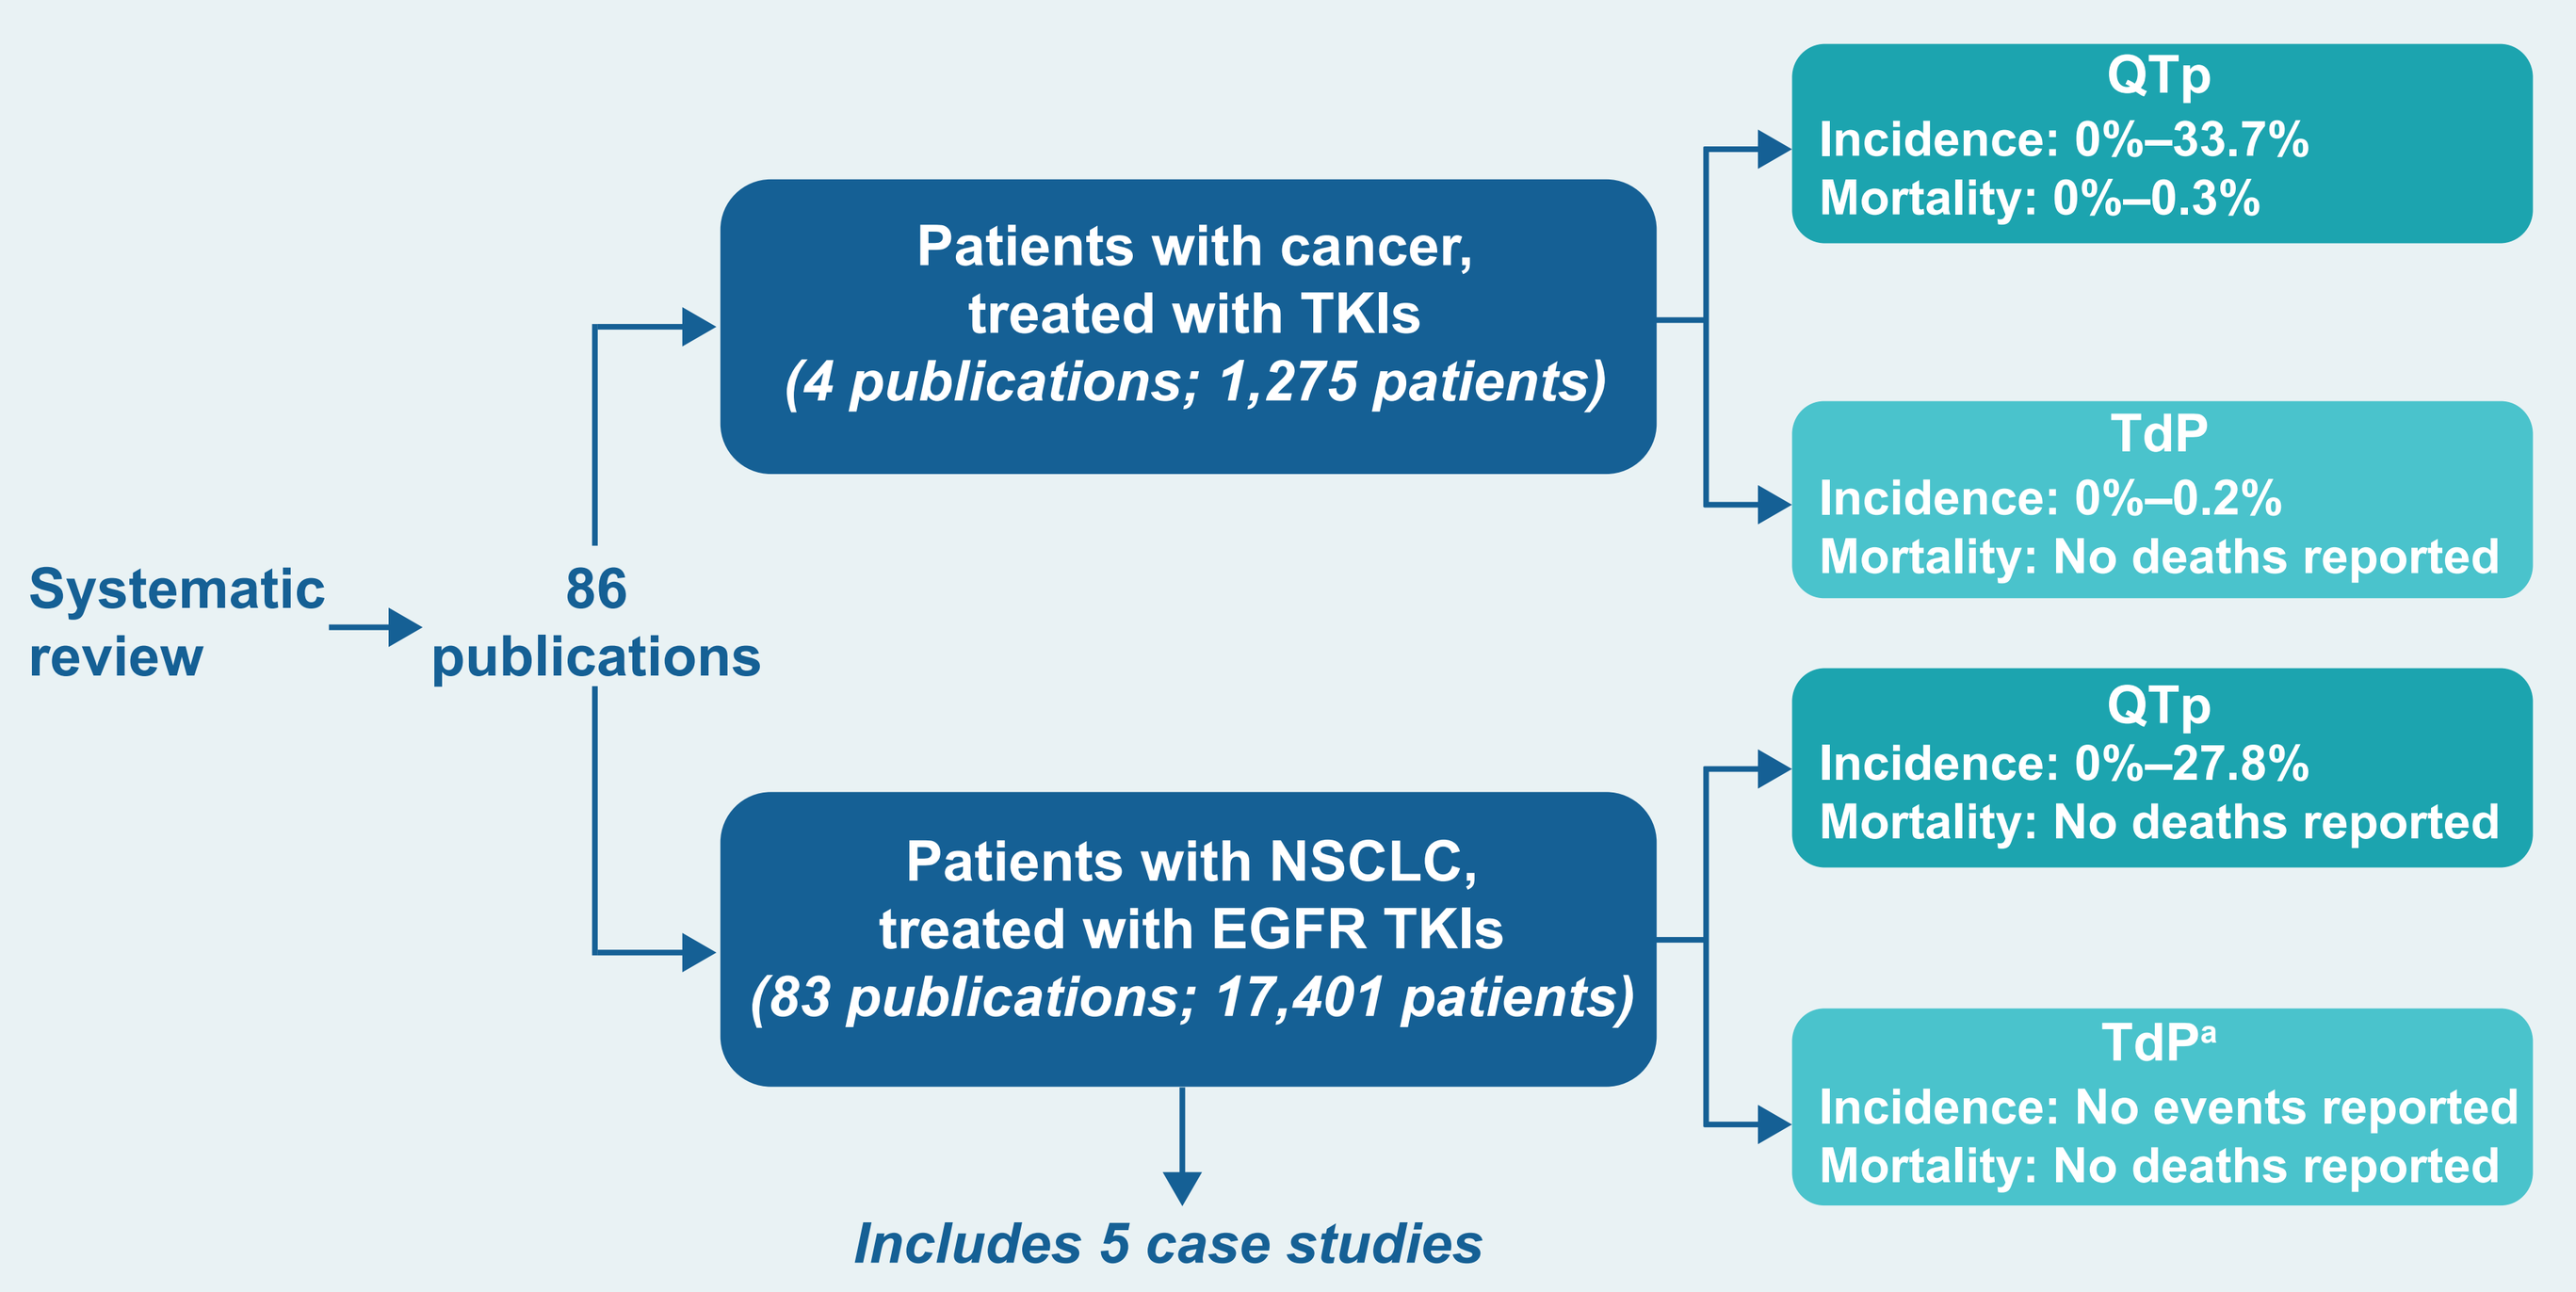
Systematic review
86 publications
Patients with cancer, treated with TKIs (4 publications; 1,275 patients)
QTp
Incidence: 0%–33.7%
Mortality: 0%–0.3%
TdP
Incidence: 0%–0.2%
Mortality: No deaths reported
Patients with NSCLC, treated with EGFR TKIs (83 publications; 17,401 patients)
QTp
Incidence: 0%–27.8%
Mortality: No deaths reported
TdP[a]
Incidence: No events reported
Mortality: No deaths reported
Includes 5 case studies

[a]NCI-CTCAE versions 3, 4, and 5 define TdP as a grade 4 QTp event, and the terminology varied; thus, TdP events may not have been described separately by investigators.

Key Takeaway Few studies reported mortality due to QTp or TdP after treatment with TKIs

Figure 4: Patients with NSCLC treated with EGFR TKIs

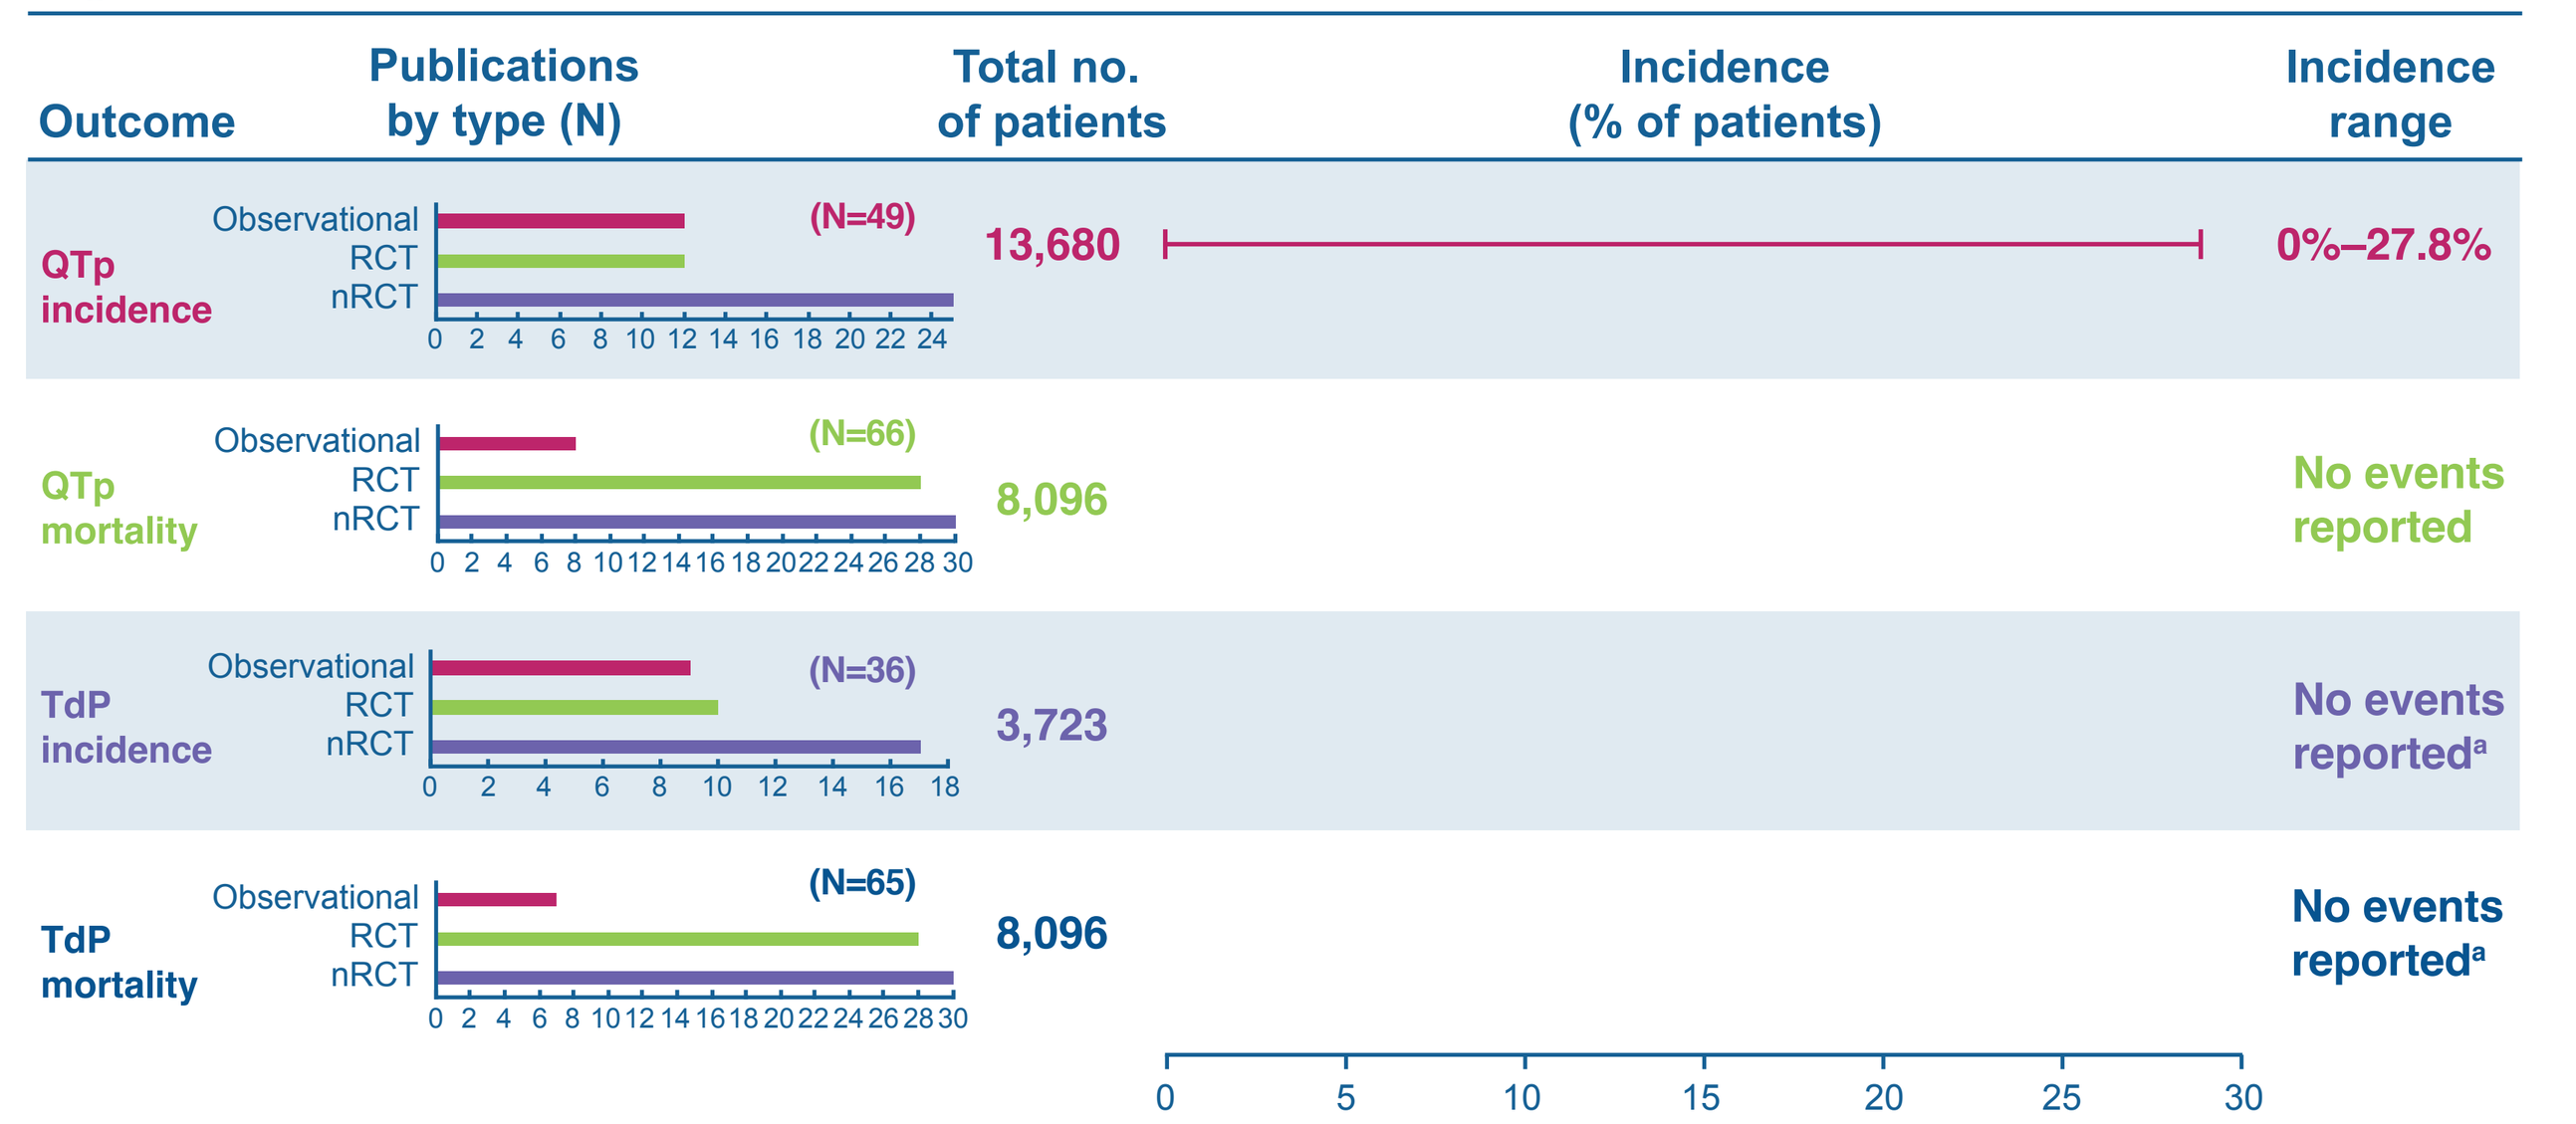
Outcome
Publications by type (N)
Total no. of patients
Incidence (% of patients)
Incidence range
QTp incidence
Observational
RCT
nRCT
(N=49)
13,680
0%–27.8%
QTp mortality
(N=66)
8,096
No events reported
TdP incidence
(N=36)
3,723
No events reported[a]
TdP mortality
(N=65)
8,096
No events reported[a]
0 5 10 15 20 25 30

[a]NCI-CTCAE versions 3, 4, and 5 define TdP as a grade 4 QTp event, and the terminology varied; thus, it is possible that TdP events were not described separately by investigators.
nRCT, nonrandomized controlled clinical trial; RCT, randomized controlled clinical trial

- Publications reporting the incidence of QTp in patients with NSCLC by EGFR TKI and reported rates are shown in **Figure 5**
 - None of the 7 studies (N=592) on icotinib reported any QTp incidence rates, and no events were reported from 1 study (N=41) on simotinib
 - 1 case report was identified that described a QTp event in a patient receiving afatinib
 - 3 case reports reported QTp events in patients receiving osimertinib
- The incidence of QTp and TdP reported for EGFR TKIs with US prescribing information is shown in **Table 1**

Figure 5: Publications reporting incidence of QTp and reported rates in patients with NSCLC by EGFR TKI

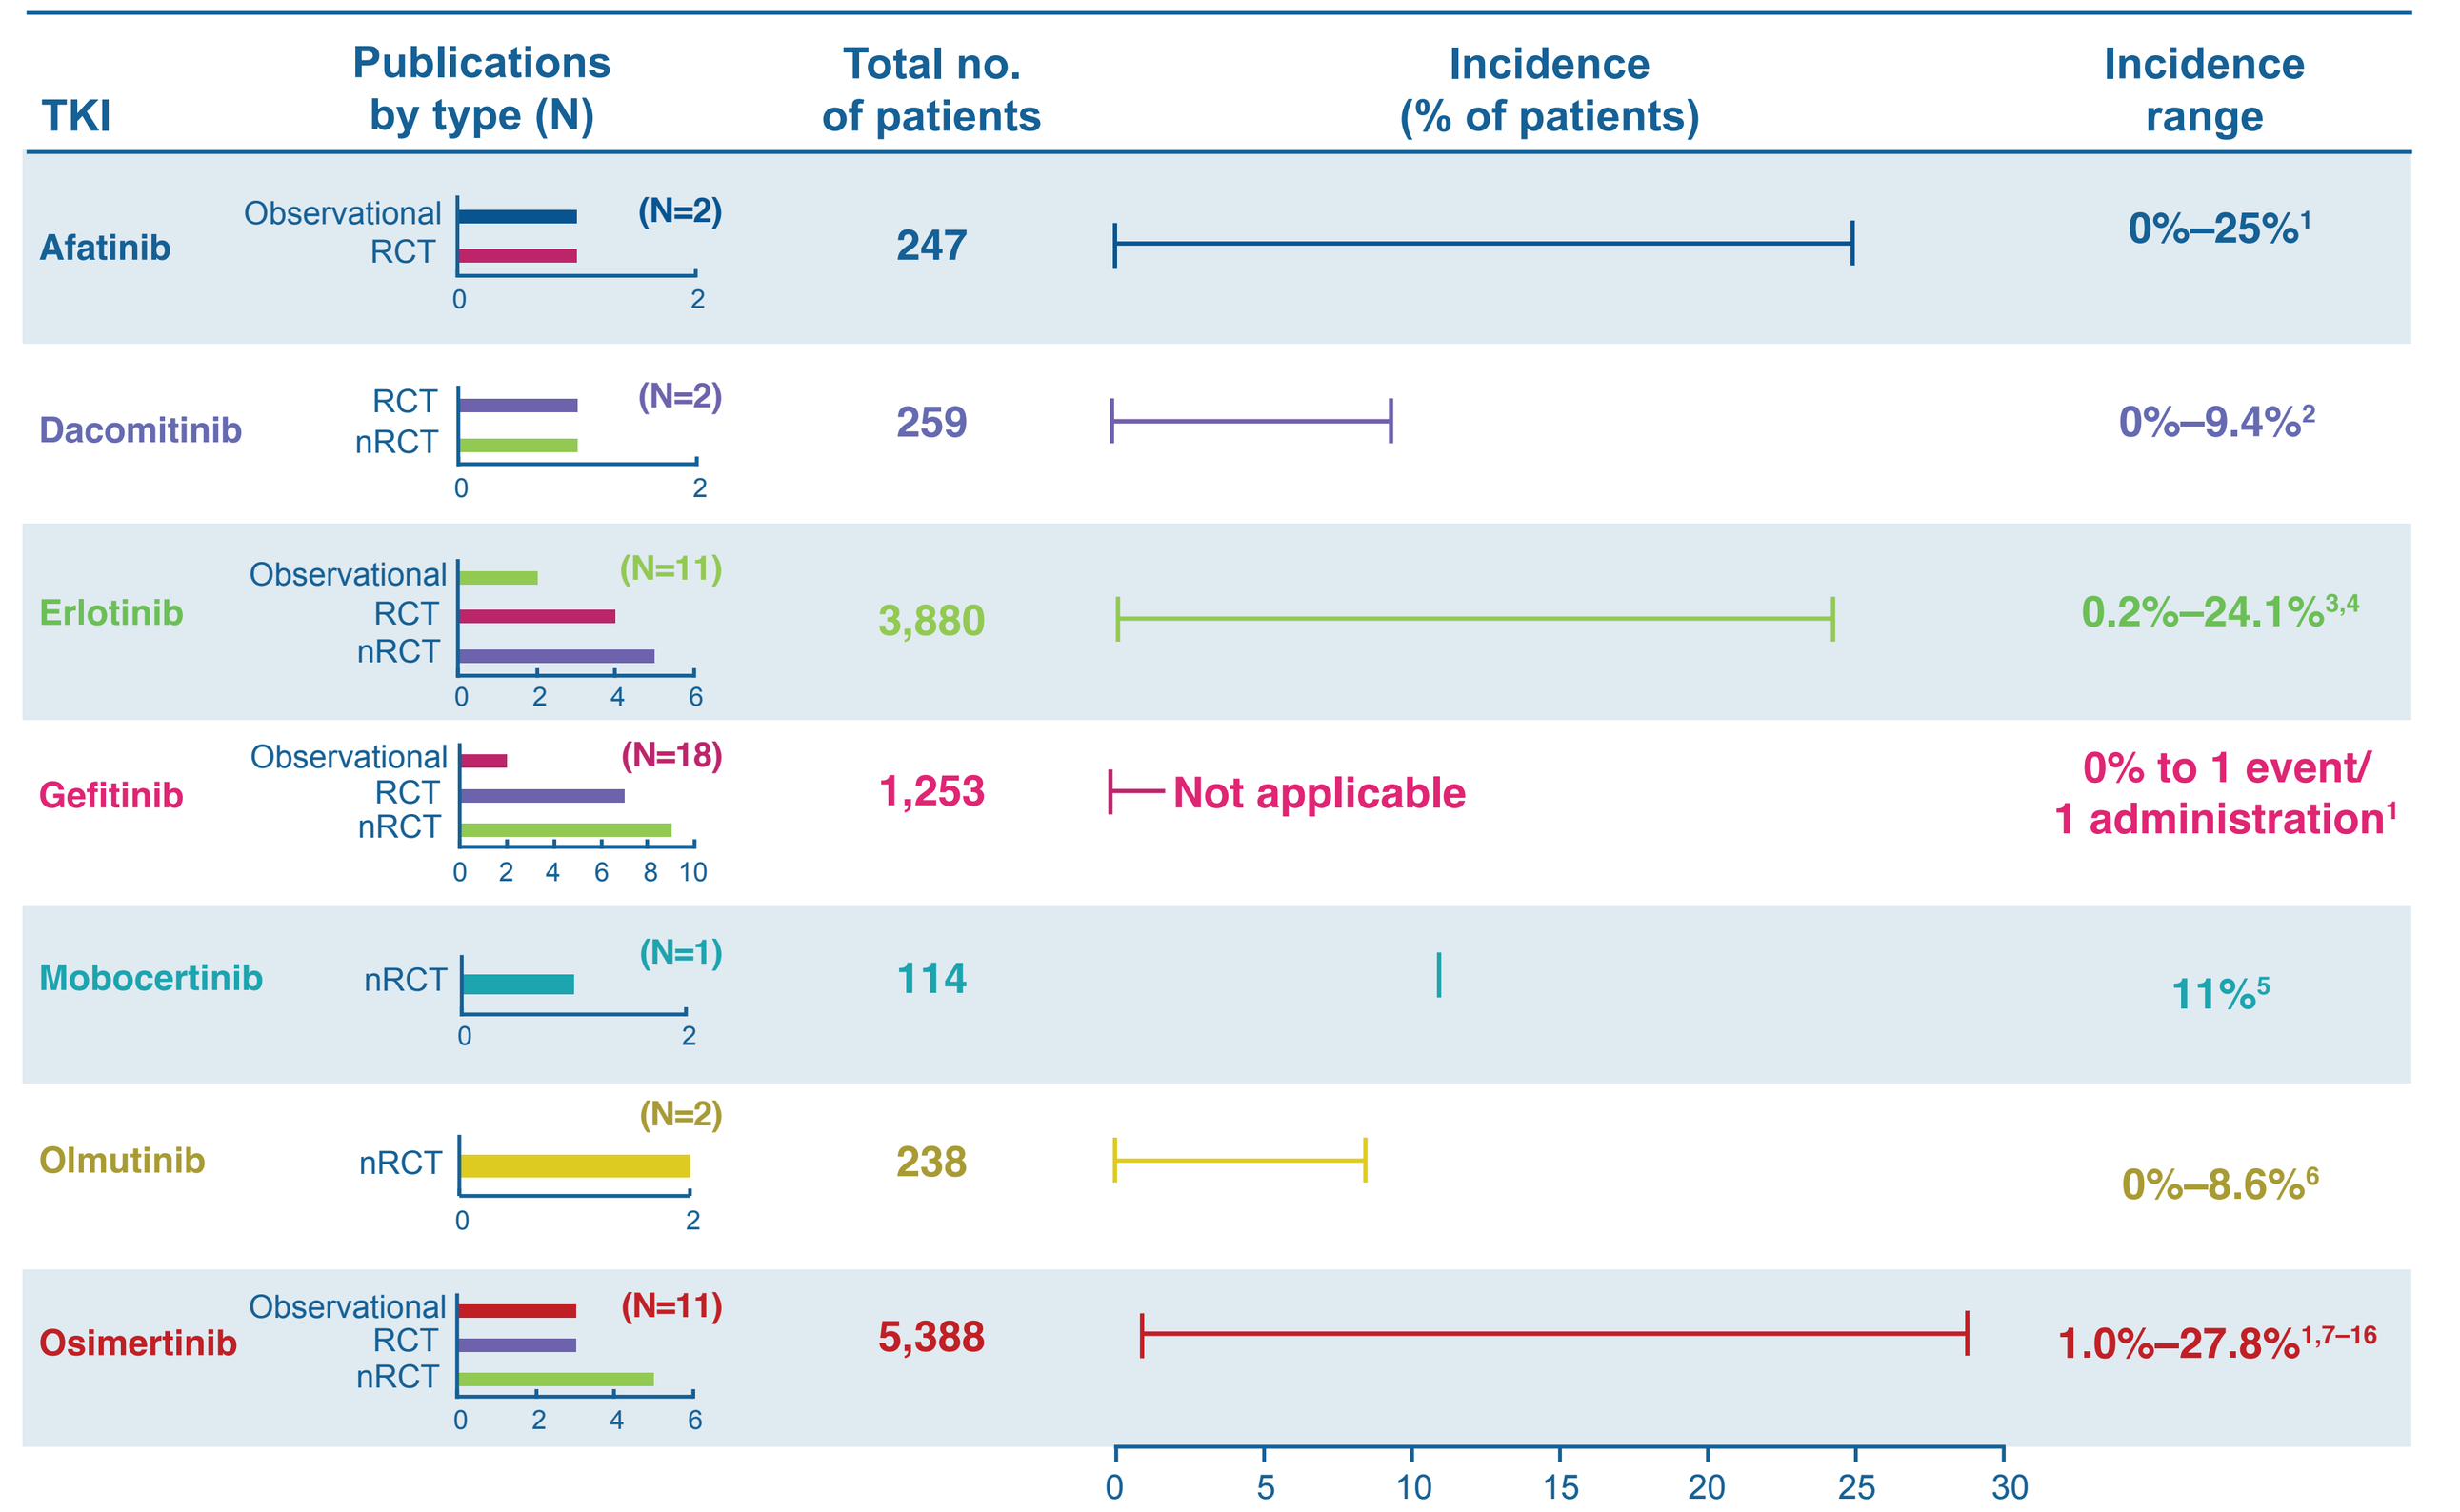
TKI
Publications by type (N)
Total no. of patients
Incidence (% of patients)
Incidence range
Afatinib
Observational
RCT
(N=2)
247
0%–25%[1]
Dacomitinib
RCT
nRCT
(N=2)
259
0%–9.4%[2]
Erlotinib
Observational
RCT
nRCT
(N=11)
3,880
0.2%–24.1%[3,4]
Gefitinib
(N=18)
1,253
Not applicable
0% to 1 event/ 1 administration[1]
Mobocertinib
nRCT
(N=1)
114
11%[5]
Olmutinib
nRCT
(N=2)
238
0%–8.6%[6]
Osimertinib
(N=11)
5,388
1.0%–27.8%[1,7–16]
0 5 10 15 20 25 30

1. Abu Rmilah AA, et al. Int J Cancer. 2020;147(11):3160–67. 2. Tan W, et al. Invest New Drugs. 2020;38(3):874–84. 3. Ehrenstein V, et al. Pharmacoepidemiol Drug Saf. 2021;30(6):758–69. 4. Natale RB, et al. J Clin Oncol. 2011;29(8):1059–66. 5. Zhou C, et al. JAMA Oncol. 2021;7(12):e214761. 6. Park K, et al. Cancer. 2021;127(9):1407–16. 7. Ramalingam SS, et al. N Engl J Med. 2020;382(1):41–50. 8. Kunimasa K, et al. JACC CardioOncol. 2020;2(1):1–10. 9. Ohe Y, et al. Jpn J Clin Oncol. 2020;50(8):909–19. 10. Goss G, et al. Lancet Oncol. 2016;17(12):1643–52. 11. Jänne PA, et al. N Engl J Med. 2015;372(18):1689–99. 12. Sequist LV, et al. Lancet Oncol. 2020;21(3):373–86. 13. Yang JC, et al. J Clin Oncol. 2017;35(12):1288–96. 14. Yu HA, et al. Clin Cancer Res. 2021;27(4):992–1002. 15. Mok TS, et al. N Engl J Med. 2017;376(7):629–40. 16. Wu YL, et al. N Engl J Med. 2020;383(18):1711–23.

Table 1: Incidence of QTp and TdP reported for EGFR TKIs in US prescribing information

Drug	QTp incidence	TdP-related information
Afatinib[5]	No large changes in the mean QTc interval (ie, >20 ms)	Not reported
Dacomitinib[6]	No large effect on QTc (ie, >20 ms) at maximum dacomitinib concentrations achieved at 45 mg oral QD (n=32)	Not reported
Erlotinib[7]	Not reported	Not reported
Gefitinib[8]	Not reported	Not reported
Mobocertinib[9]	QTc interval >500 ms: 1.2% Change-from-baseline QTc interval >60 ms: 11% (n=250[a])	Grade 4 TdP: 0.4% (1 patient) (n=250)
Osimertinib[10]	QTc >500 ms: 0.8% Increase-from-baseline QTc >60 ms: 3.1% (n=1479)	No QTc-related arrhythmias were reported (n=1479)

[a]Subset of the pooled mobocertinib safety population that had scheduled and unscheduled electrocardiograms. Follow-up time was not reported for this subset of 250 patients; however, for all 256 patients in the pooled safety population, 48% were exposed for ≥6 months and 12% were exposed >1 year.
Note: Almonertinib and icotinib are approved in China but not in the United States; olmutinib development was discontinued in 2018; simotinib was in development at time of review.
QD, once daily; QTc, QT corrected for heart rate

Limitations

- Notable strengths of this report include:
 - Use of a prespecified protocol, search criteria, and abstraction sheet, as well as the wide breadth of literature evaluated
 - Being the first systematic review to investigate the incidence and mortality of QTp and TdP associated with EGFR TKIs
- Limitations and challenges in identifying relevant data for this report include:
 - Only covered publications in the English language
 - Treatment guidelines and drug availability vary by region, complicating comparisons among studies
 - Some measures of incidence may actually reflect prevalence, incident events, or worsening of events but not an incident condition
 - Possible misclassification of events
 - Follow-up time varied across studies and even within different treatment arms of a single study
 - Different outcome definitions/criteria and different versions of the National Cancer Institute's Common Terminology Criteria for Adverse Events were used across studies
 - NCI-CTCAE versions 3, 4, and 5 define TdP as a grade 4 QTp event, and the terminology varied; thus, TdP events may not have been described separately by investigators. This could potentially explain why no TdP events or deaths were recorded in some cases

Conclusions

- In patients with cancer receiving TKI treatment, the incidence of QTp was variable, ranging from 0% to 37.7%; only 1 study reported QTp mortality rate of 0.3%
- In patients with NSCLC receiving EGFR TKIs, QTp events were observed in approximately 0%–27.8% of patients, with no deaths reported
 - No TdP events and no deaths due to TdP were reported in observational studies or clinical trials over the search period; however, because NCI-CTCAE versions 3–5 defined TdP as a grade 4 QTp event, TdP events may not have been reported separately by investigators
- Health care providers should monitor patients for signs of cardiotoxicity, specifically related to changes in heart rhythm, consistent with prescribing information for EGFR TKIs
- Potential future directions include the development of standardized definitions of QTp and methods to improve monitoring for QTp and TdP in patients with cancer

References

1. Arunachalam K, et al. J Clin Med Res. 2018;10(5):384–90.
2. Anderson HN, et al. J Emerg Med. 2018;54(1):8–15.
3. Kunimasa K, et al. JACC: Cardiooncology. 2020;2(1):1–10.
4. Kloth JS, et al. Br J Cancer. 2015;112(6):1011–16.
5. Gilotrif [package insert]. Ridgefield, CT: Boehringer Ingelheim Pharmaceuticals, Inc.; 2022.
6. Vizimpro [package insert]. New York, NY: Pfizer Inc.; 2020.
7. Tarceva [package insert]. Northbrook, IL: OSI Pharmaceuticals, LLC; 2016.
8. Iressa [package insert]. Wilmington, DE: AstraZeneca Pharmaceuticals LP; 2021.
9. Exkivity [package insert]. Lexington, MA: Takeda Pharmaceuticals America, Inc.; 2023.
10. Tagrisso [package insert]. Wilmington, DE: AstraZeneca Pharmaceuticals; 2023.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Corey Burgin, PhD, of Peloton Advantage, LLC, an OPEN Health company; funded by Takeda Development Center Americas, Inc., Lexington, MA, USA; and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. Ann Intern Med 2022;175:1298–1304). This literature review was sponsored by Takeda Development Center Americas, Inc.

Disclosures

BK and **BC:** Employment with Takeda. **KI:** Consulted on this project through CERobs Consulting, LLC; Takeda Pharmaceuticals contracted with CERobs Consulting, LLC, a consulting firm with focus on real-world evidence, outcomes research, epidemiology, and clinical outcome assessments, including patient-reported outcomes. **KR** and **NRO:** Consulted on this project through a collaboration between the University of North Carolina at Chapel HIll and CERobs Consulting, LLC; Takeda Pharmaceuticals contracted with CERobs Consulting, LLC, a consulting firm with focus on real-world evidence, outcomes research, epidemiology, and clinical outcome assessments, including patient-reported outcomes. **WS** and **EF:** Employment with Takeda.

For an e-Print, scan this QR code.
Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the World Conference on Lung Cancer (WCLC), September 9–12, 2023, Singapore

Email for questions or comments: Bilal.Khokhar@takeda.com